Analysis of stroma and immune-related gene expression patterns during breast cancer (BC) progression

1706PD
ESMO Congress 2017

Luciana Molinero[1], Joan Albanell[2,3,4], Hartmut Koeppen[1], Eduardo Martínez de Dueñas[2,5], Dan Halligan[6], Ángel Guerrero-Zotano[2,7], Jose Ignacio Chacón[2,8], Ramón Pérez[2,9], Silvia Antolín[2,10], Isabel Blancas[2,11], Montserrat Muñoz[2,12], Amparo Oltra[2,13], María Helena López-Ceballos[2,14], María Sánchez-Aragó[2], Rosalía Caballero[2], Eva Carrasco[2], Ana María González-Angulo[15], Ana Lluch[2,3,16], Elizabeth A. Mittendorf[15], Federico Rojo[2,3,17]

[1]Genentech, San Francisco, CA; [2]GEICAM (Spanish Breast Cancer Group), Madrid, Spain; [3]Centro de Investigación Biomédica en Red de Oncología, CIBERONC-ISCIII, Spain; [4]Hospital del Mar, Barcelona, Barcelona, Spain;[5]Hospital Provincial de Castellón, Castellón, Spain; [6]Fios Genomics, Edinburgh, UK; [7]Instituto Valenciano de Oncología, Valencia, Spain; [8]Hospital Virgen de la Salud, Toledo, Spain; [9]Hospital Universitario Quirón de Madrid, Madrid, Spain;[10]Complejo Hospitalario Universitario A Coruña; [11]Hospital Clínico San Cecilio, Granada, Spain;[12]Hospital Clinic i Provincial, Barcelona, Spain; [13]Hospital Virgen de los Lirios, Alicante, Spain; [14]Hospital San Pedro de Alcántara, Cáceres, Spain; [15]The University of Texas MD Anderson Cancer Center, Houston, TX, [16]Hospital Clínico Universitario de Valencia, Valencia, Spain, [17]Fundación Jiménez Díaz, Madrid, Spain.

GEicam
spanish breast cancer group

INTRODUCTION

- ✓ Despite accumulating evidences sustaining that immune cells are generic constituents of tumors (1), the role that the immune system plays in tumorigenesis is still a matter of debate.
- ✓ Characterization of the immune phenotype of tumors during progression could aid at developing patient-tailored therapy strategies.
- ✓ Here, we sought to **identify differences in the expression pattern of immune-related genes comparing paired samples of primary breast tumors and metastasis from patients participant in the GEICAM/2009-03 (ConvertHER) study.**

OBJECTIVES

- ✓ To characterize the **immune phenotype during breast cancer progression** in terms of:
 - ✓ **PD-L1 expression** and
 - ✓ **Immune gene expression** analysis.

MATERIALS AND METHODS

- ✓ We analyzed in **44** matched pairs, **1** primary and **1** metastatic non-matched tissues from **46** patients participant in the ConvertHER study, **PD-L1 expression** by immunohistochemistry (IHC) by using a specific antibody for tumor and immune cells detection (2) **(Figure 1).**

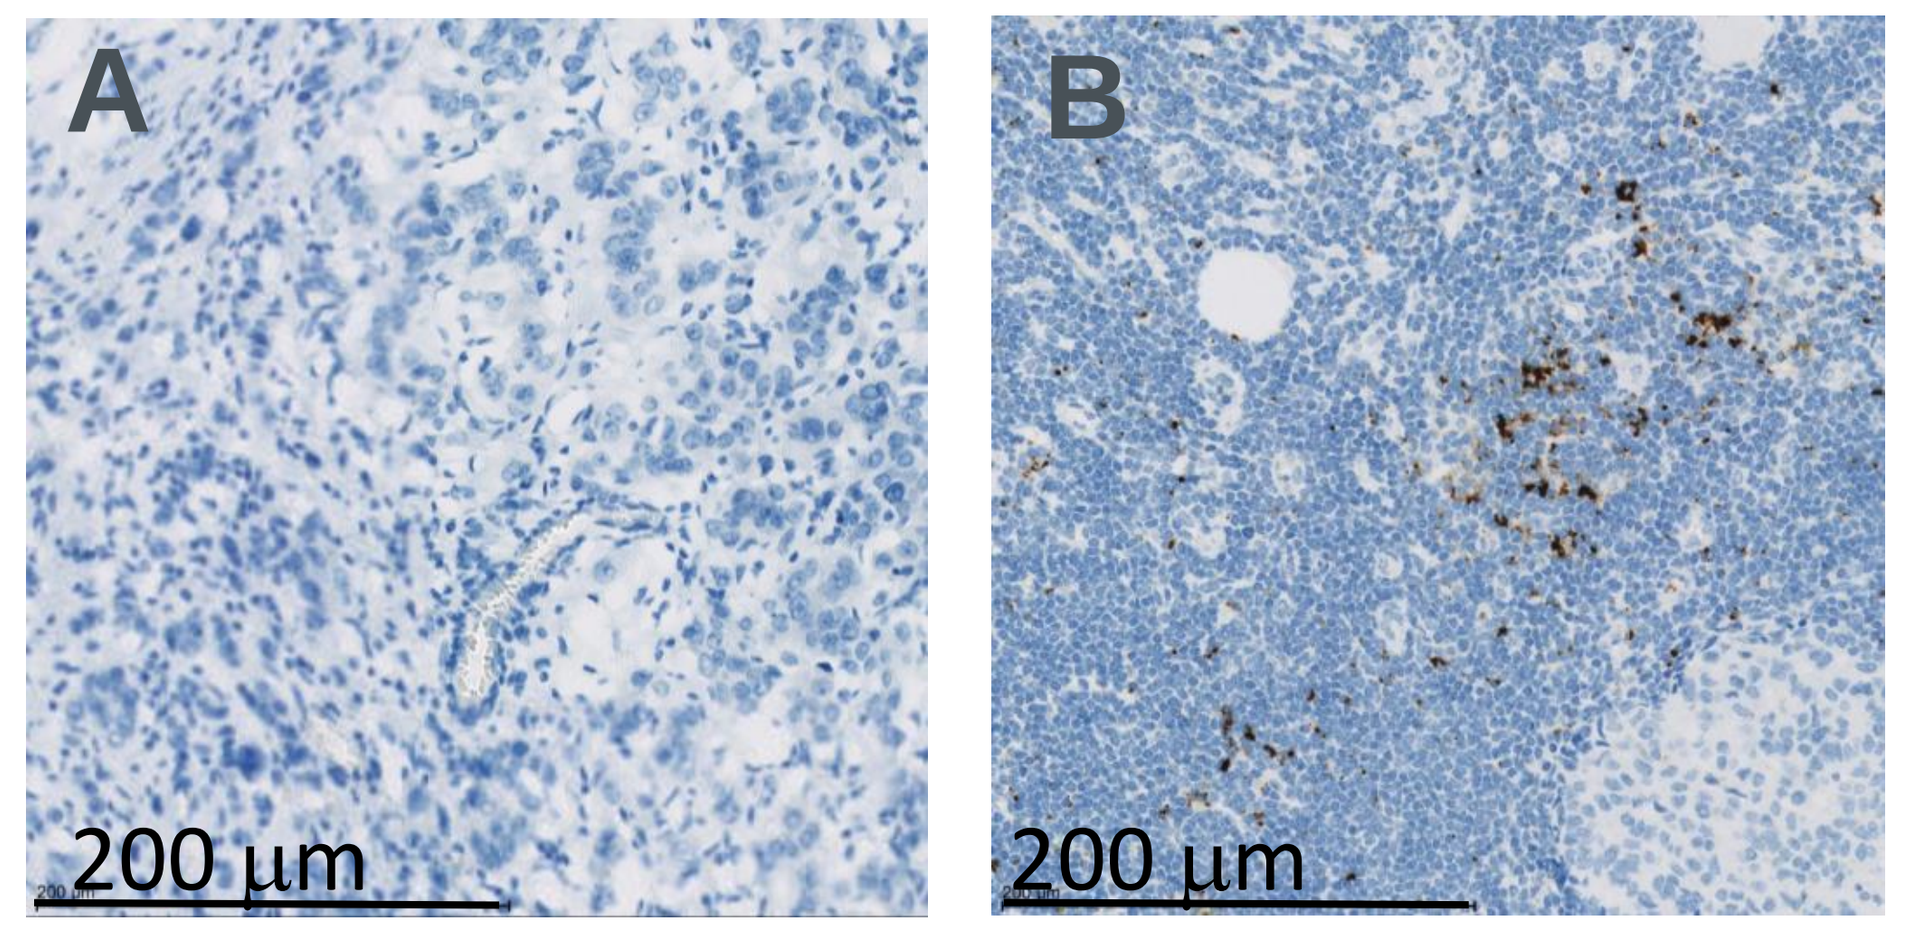
A
B
200 µm
200 µm

Figure 1. Representative IHC for PD-L1 antibody (SP142 clone) in breast cancer tissue. (A). Primary BC with less than 1% of tumor infiltrating immune cells (IC) expressing PD-L1 (PD-L1 IC<1%) **(B)** Primary BC with more than 1% IC expressing PD-L1 (PD-L1 IC≥1%).

- ✓ Differential gene expression between **60 matched primary/metastases pairs** was analyzed in a **customized nanostring** (enriched in immune-related genes). .
- ✓ Significant features (p-value <0.05) were assessed for functional enrichment of KEGG pathways and GO terms.

RESULTS

- ✓ **Prevalence of PD-L1 in immune infiltrating cells (IC) was lowest in ER+HER2- specimens (Figure 2).** PDL1 expression in tumor cells (TC) was observed in 1/90 samples.
- ✓ **No significant changes in PD-L1 IC were observed between matched pairs (Figures 3 and 4).**

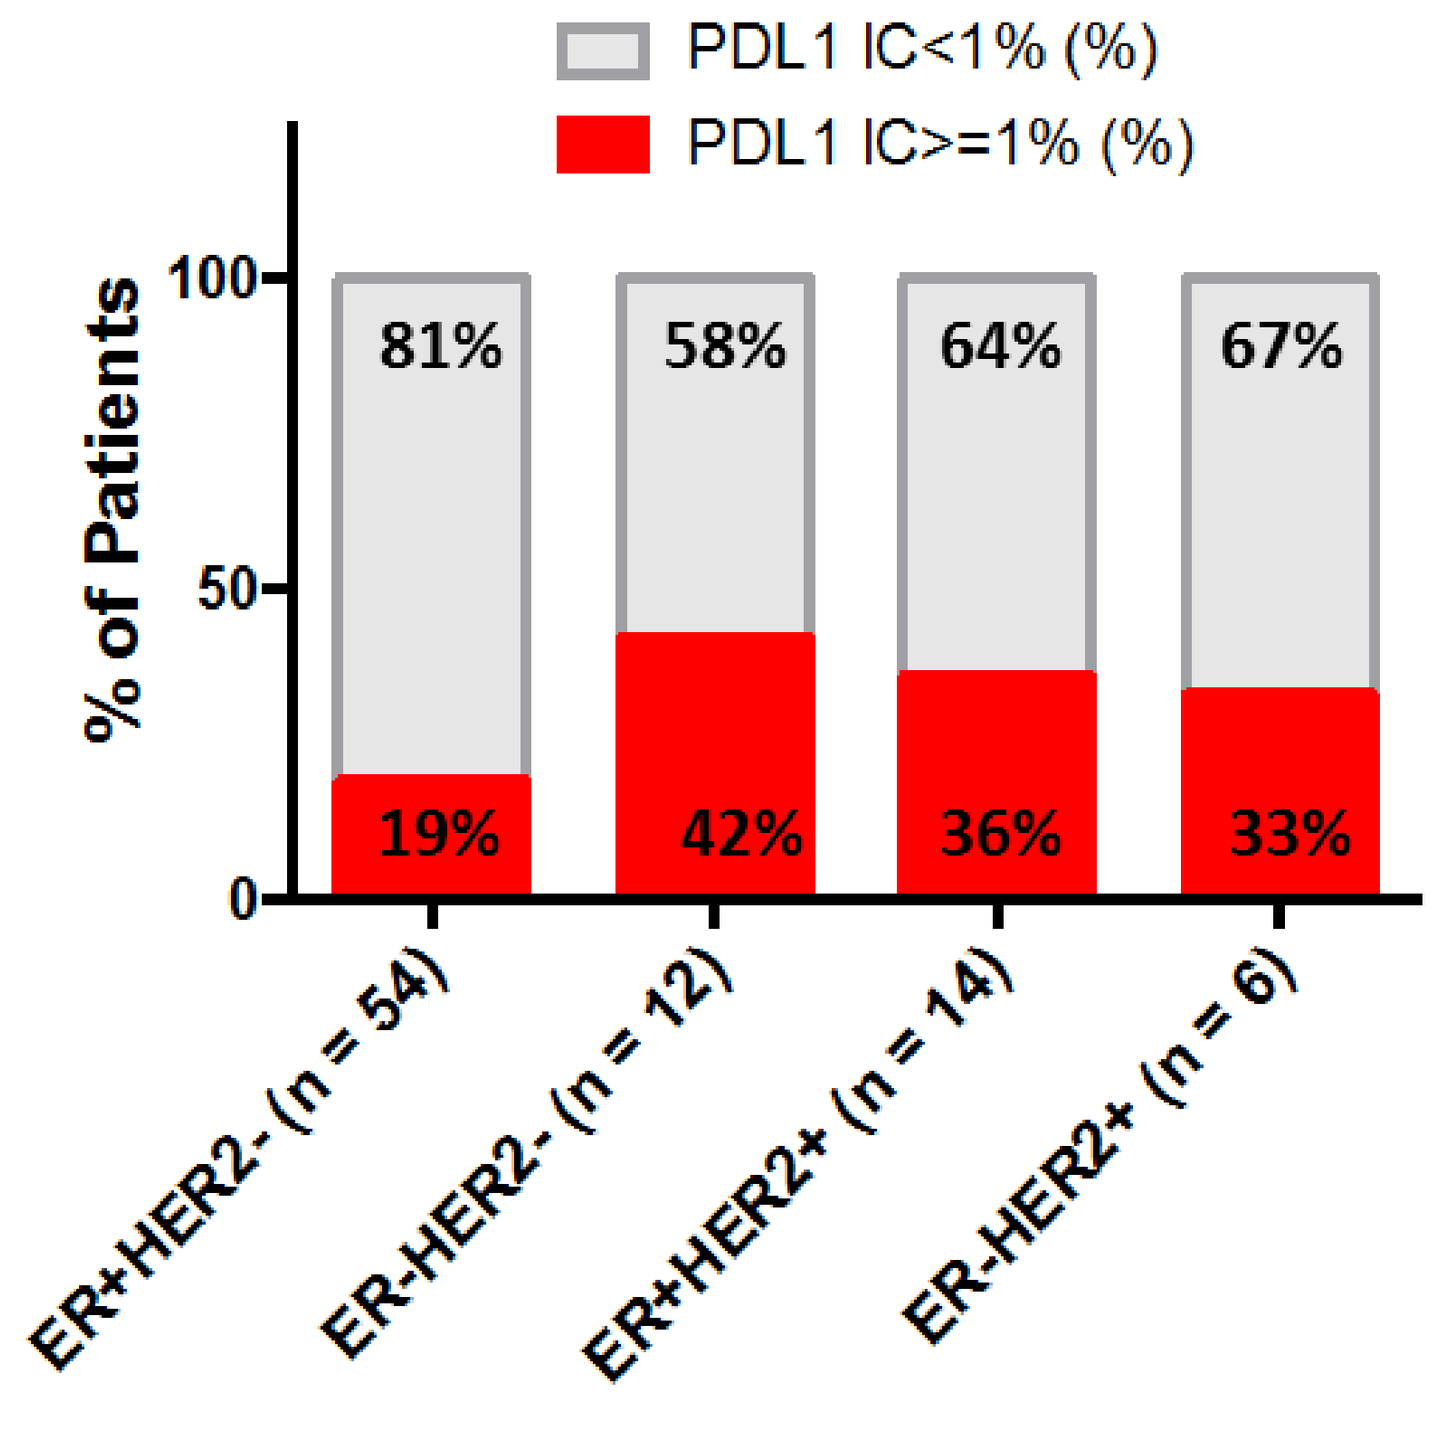
PDL1 IC<1% (%)
PDL1 IC>=1% (%)
% of Patients
100
50
0
81%
58%
64%
67%
19%
42%
36%
33%
ER+HER2- (n = 54)
ER-HER2- (n = 12)
ER+HER2+ (n = 14)
ER-HER2+ (n = 6)

Figure 2. PD-L1 IC by BC Subtype

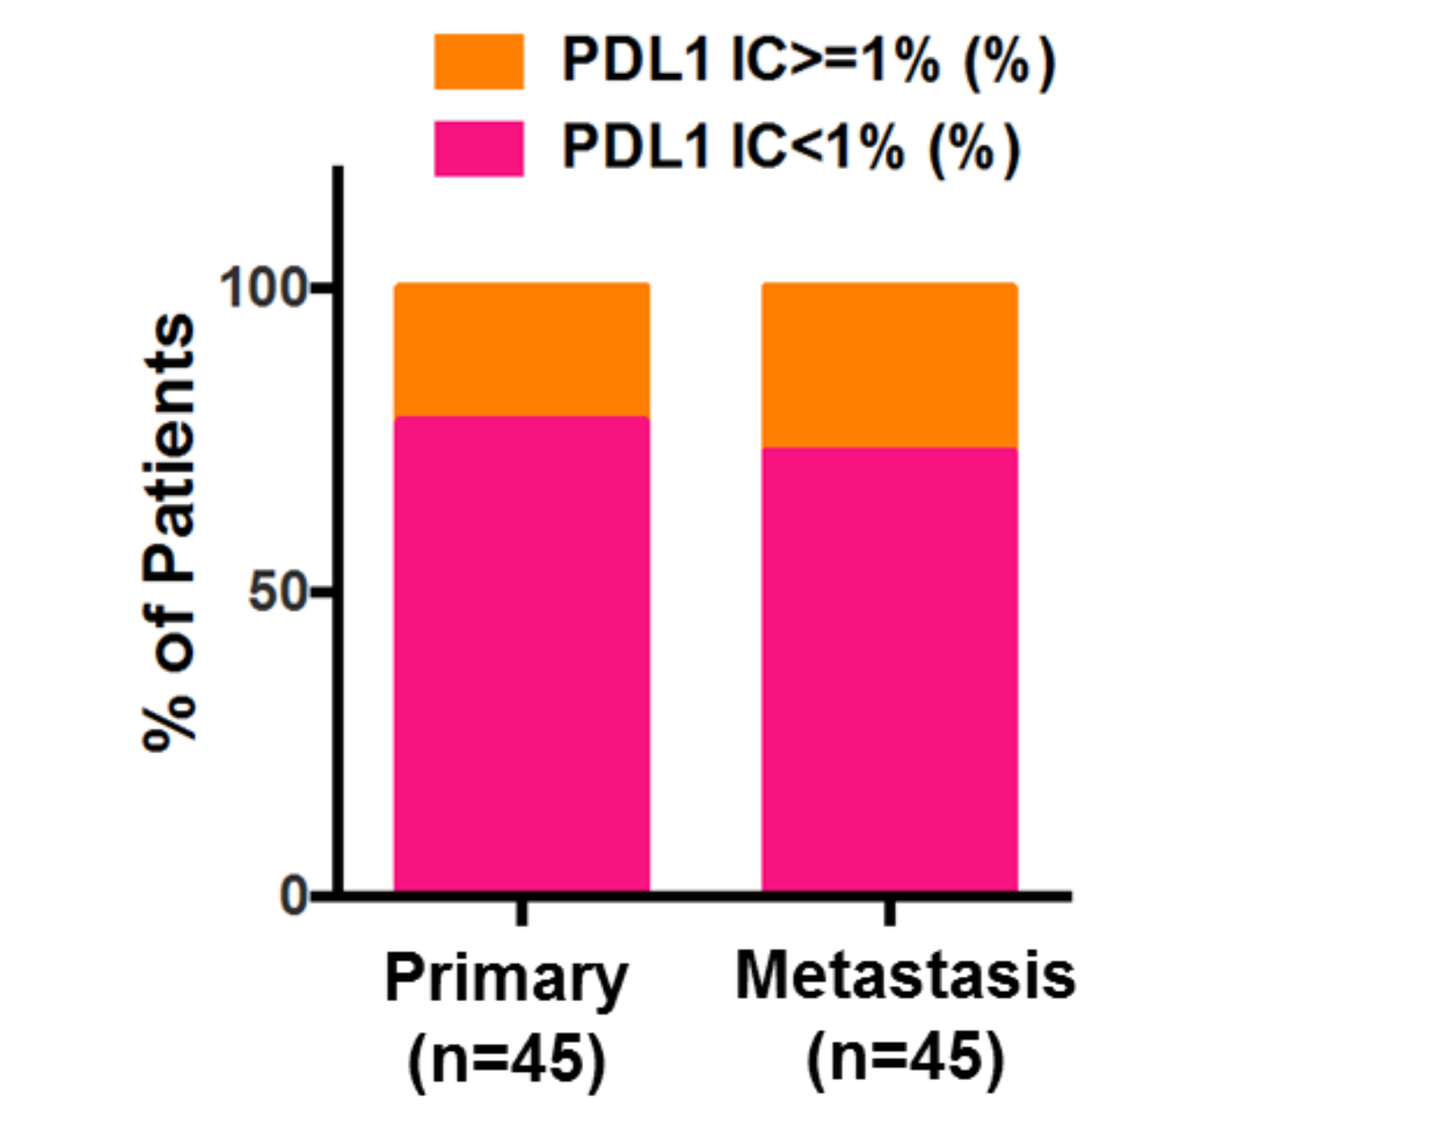
PDL1 IC>=1% (%)
PDL1 IC<1% (%)
% of Patients
100
50
0
Primary (n=45)
Metastasis (n=45)

Figure 3. Prevalence PD-L1 IC in primary and metastasis from 44 matched pairs, 1 primary and 1 metastatic non-matched tissues.

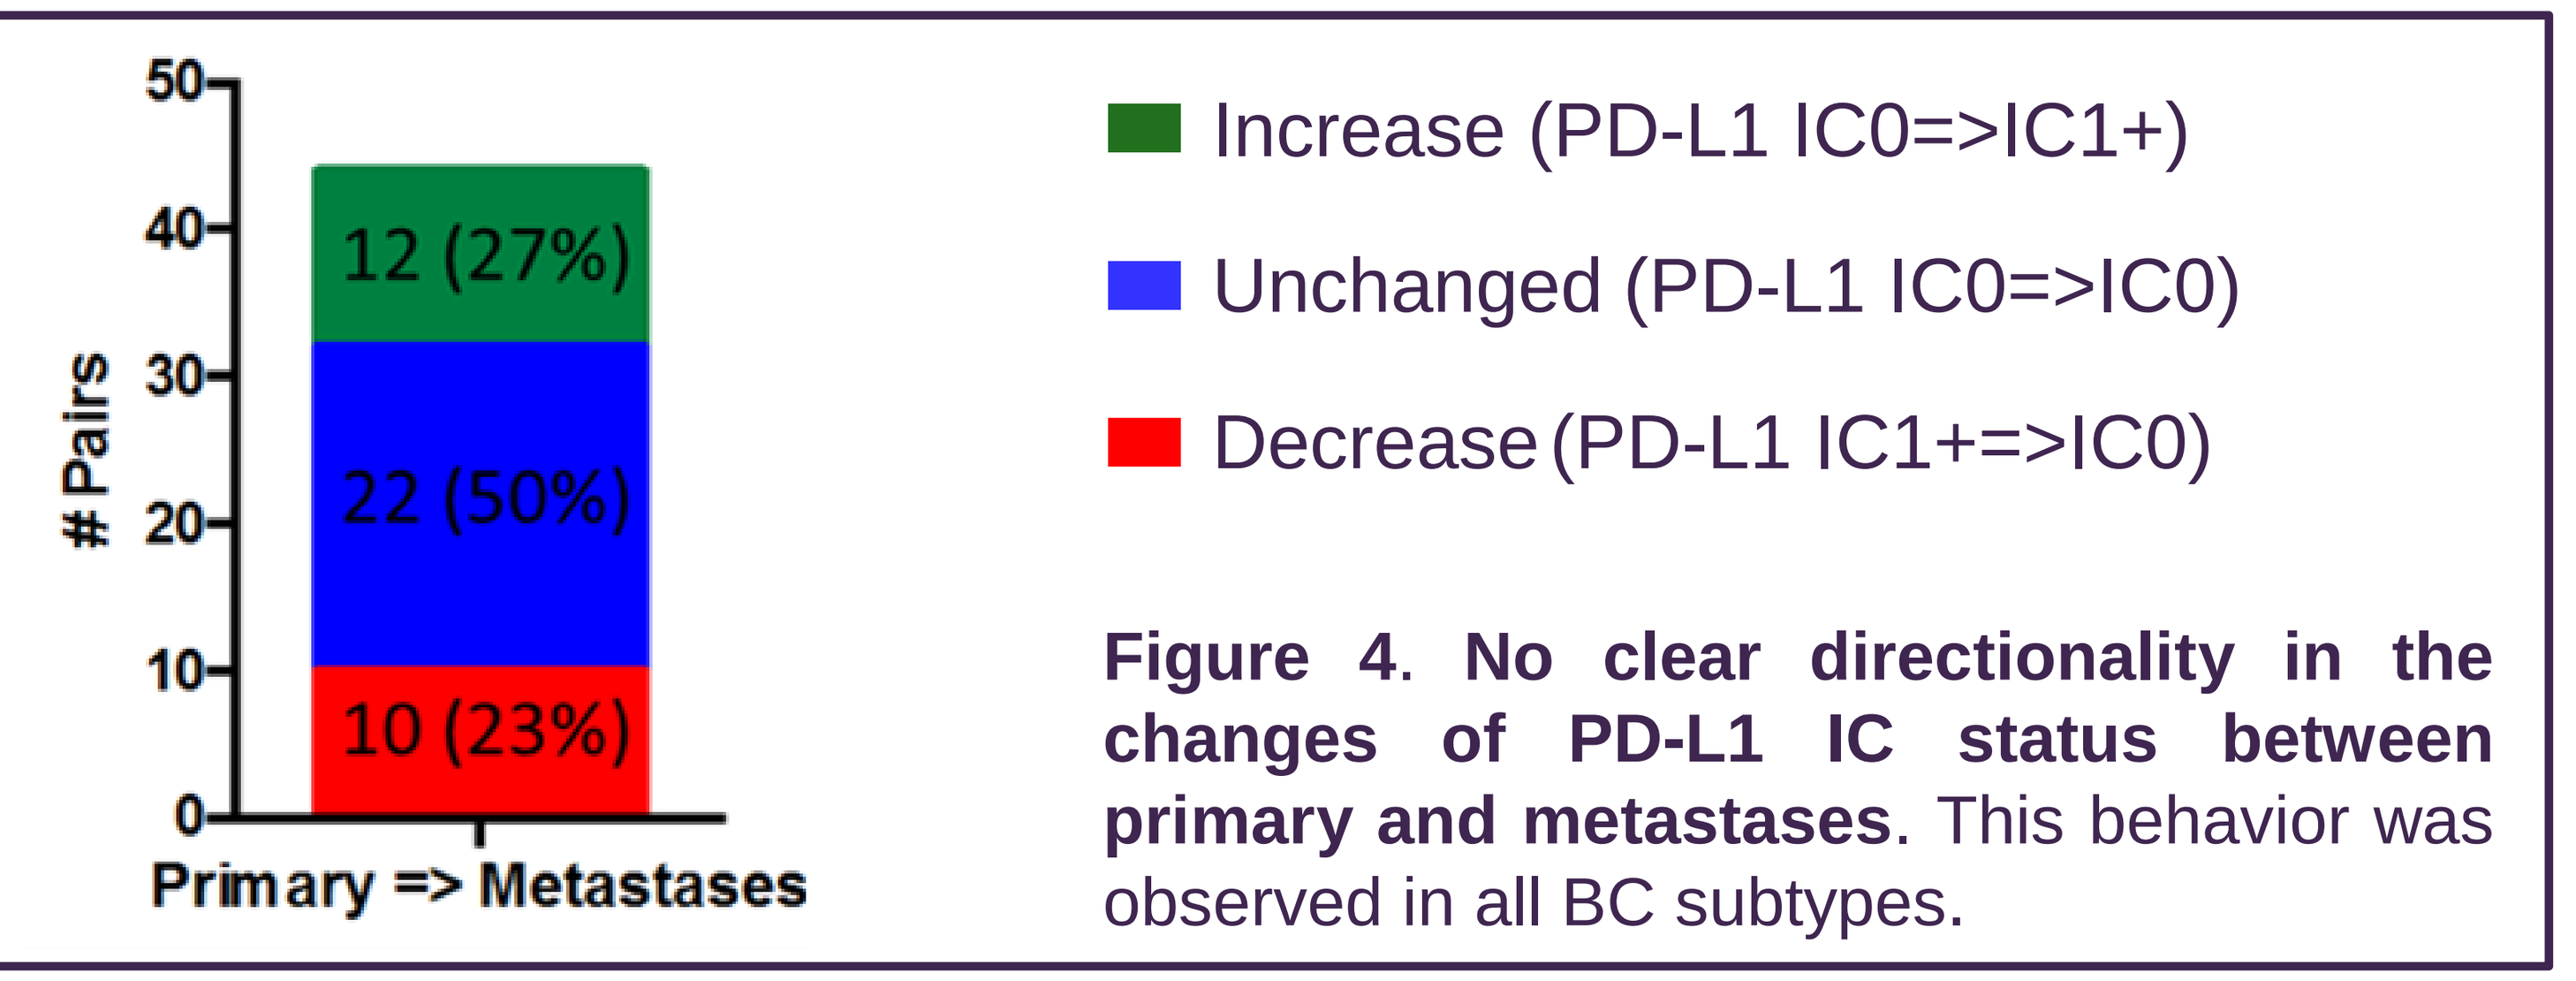
Pairs
50
40
30
20
10
0
12 (27%)
22 (50%)
10 (23%)
Primary => Metastases
Increase (PD-L1 IC0=>IC1+)
Unchanged (PD-L1 IC0=>IC0)
Decrease (PD-L1 IC1+=>IC0)

Figure 4. No clear directionality in the changes of PD-L1 IC status between primary and metastases. This behavior was observed in all BC subtypes.

- ✓ **No changes** in pre-specified immune signatures were observed **(Figure 5).**
- ✓ Metastases had decreased Notch pathway, innate inflammation and TGFb-activated fibroblasts signatures **(Figure 6).**

60 BC pairs analyzed by nanostring

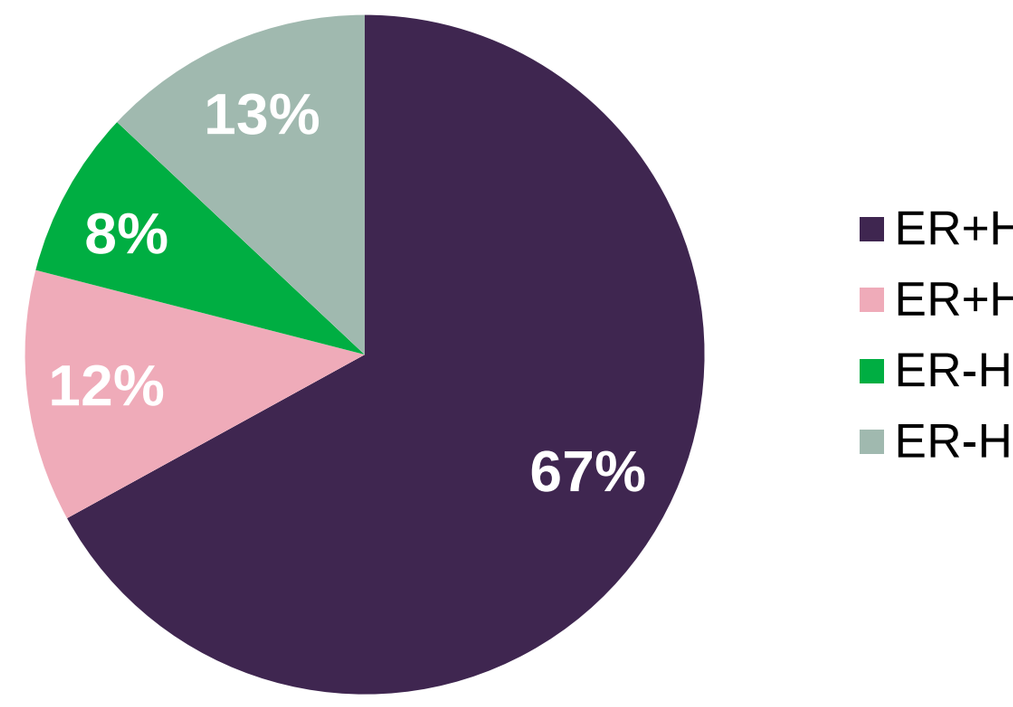
67%
12%
8%
13%
ER+HER2-
ER+HER2+
ER-HER2+
ER-HER2-

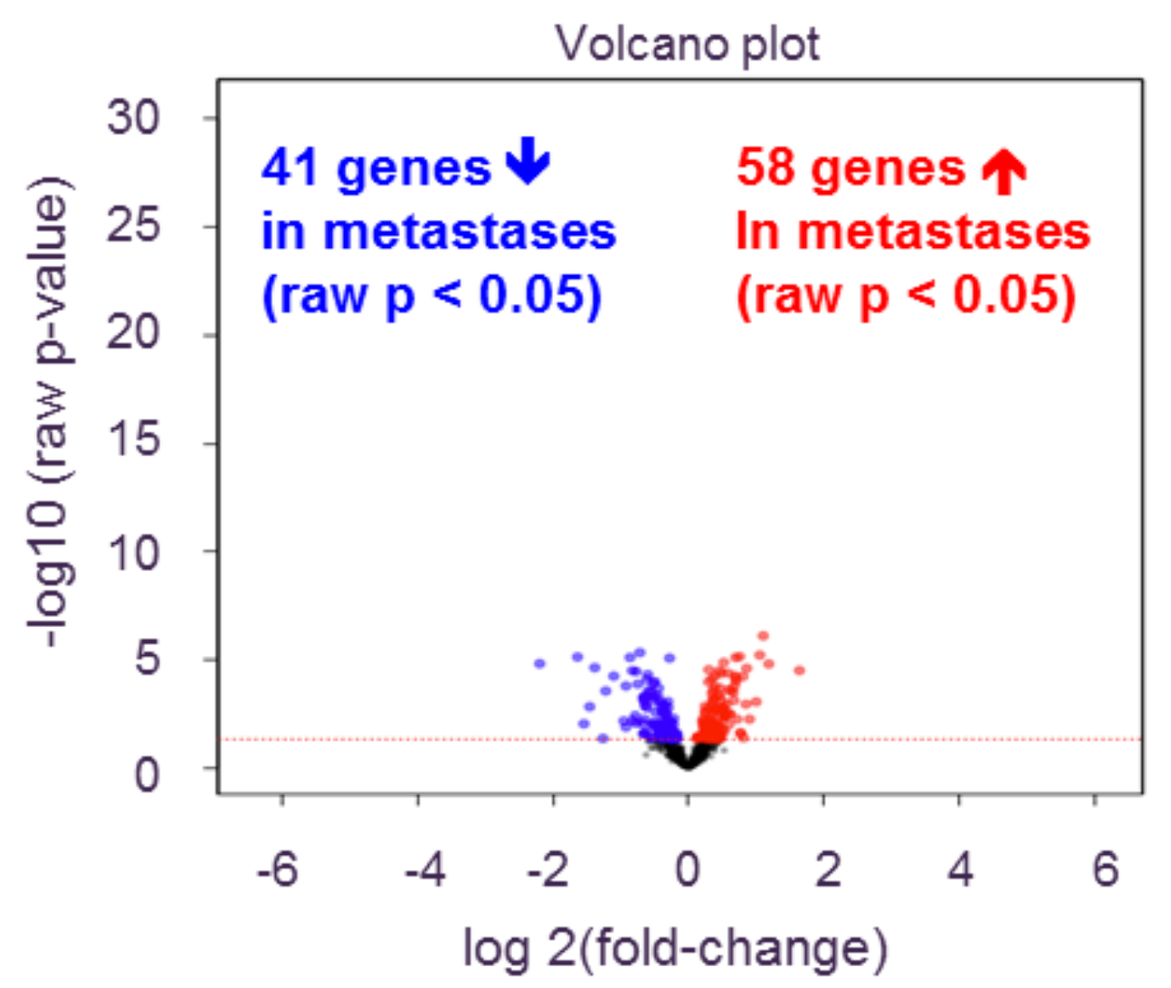
Volcano plot
41 genes in metastases (raw p < 0.05)
58 genes In metastases (raw p < 0.05)
-log10 (raw p-value)
log 2(fold-change)

Most Downregulated Genes			Most Downregulated Genes		
Gene	logFC	P.Value	Gene	logFC	P.Value
KRT17	-2.198	1.52E-05	S100A8	1.643	3.13E-05
LTF	-1.638	7.49E-06	ARG1	1.189	1.57E-05
KRT5	-1.543	0.009188	TDO2	1.107	7.83E-07
NA	-1.458	0.001489	FERMT1	1.057	6.11E-06
FOS	-1.383	2.33E-05	FGFR4	1.003	0.0008813
KRT14	-1.262	0.04378	CCL18	0.9036	0.005632
MMP7	-1.221	0.0002788	LRP4	0.8613	2.50E-05
MMP11	-1.103	5.78E-05	PRAME	0.8519	0.001133
PGR	-0.9526	0.006726	CXCL13	0.8135	0.04031
CCL19	-0.9223	0.01304	MRC1	0.8081	5.97E-05
MME	-0.9209	0.0001633	S100A9	0.773	0.0244
FGF1	-0.8617	7.95E-06	LCN2	0.7716	0.02466
MIA	-0.8282	3.18E-05	CDK6	0.7576	7.19E-06
SFRP1	-0.8197	0.006552	NCAM1	0.721	0.005617
AREG	-0.7842	0.003816	CR1	0.7051	9.55E-05
CXCL9	-0.7699	0.008736	GGH	0.7035	6.99E-05

Figure 5. Gene changes in primary vs metastatic matched pairs

Name	Pvalue
SMAD protein signal transduction	0.00271869
negative regulation of protein autophosphorylation	0.00271869
mature B cell differentiation involved in immune response	0.00271869
mature B cell differentiation	0.00271869
positive regulation of interferon-alpha production	0.00271869
MDA-5 signaling pathway	0.00271869
antimicrobial humoral response	0.00052652
antibacterial humoral response	0.00788276
regulation of cardiac muscle contraction	0.00788276
cardiac muscle contraction	0.00788276
regulation of striated muscle contraction	0.00788276
release of sequestered calcium ion into cytosol by sarcoplas	0.00788276
sarcoplasmic reticulum calcium ion transport	0.00788276
cytoplasmic pattern recognition receptor signaling pathway	0.00788276
Ras GTPase activator activity	0.00241104
response to mineralocorticoid	0.00244307
membrane depolarization	0.00244307
insulin receptor binding	0.01511404
insulin-like growth factor binding	0.01511404
Rho GTPase activator activity	0.01511404
positive regulation of lipid catabolic process	0.01523899
positive regulation of steroid biosynthetic process	0.01523899
positive regulation of steroid metabolic process	0.01523899
response to corticosterone	0.01523899
negative regulation of Ras protein signal transduction	0.01523899
negative regulation of small GTPase mediated signal transd	0.01523899
axon regeneration	0.01523899
negative regulation of myeloid cell apoptotic process	0.01523899
neuron projection regeneration	0.01523899
negative regulation by host of viral transcription	0.01523899
negative regulation of viral transcription	0.01523899
regulation of protein autophosphorylation	0.01523899
regulation of monocyte differentiation	0.01523899
glial cell migration	0.01523899
positive regulation of type I interferon-mediated signaling	0.01523899
macrophage apoptotic process	0.01523899
cellular response to dsRNA	0.01523899

Table 1. GO-enriched pathways downregulated in metastases

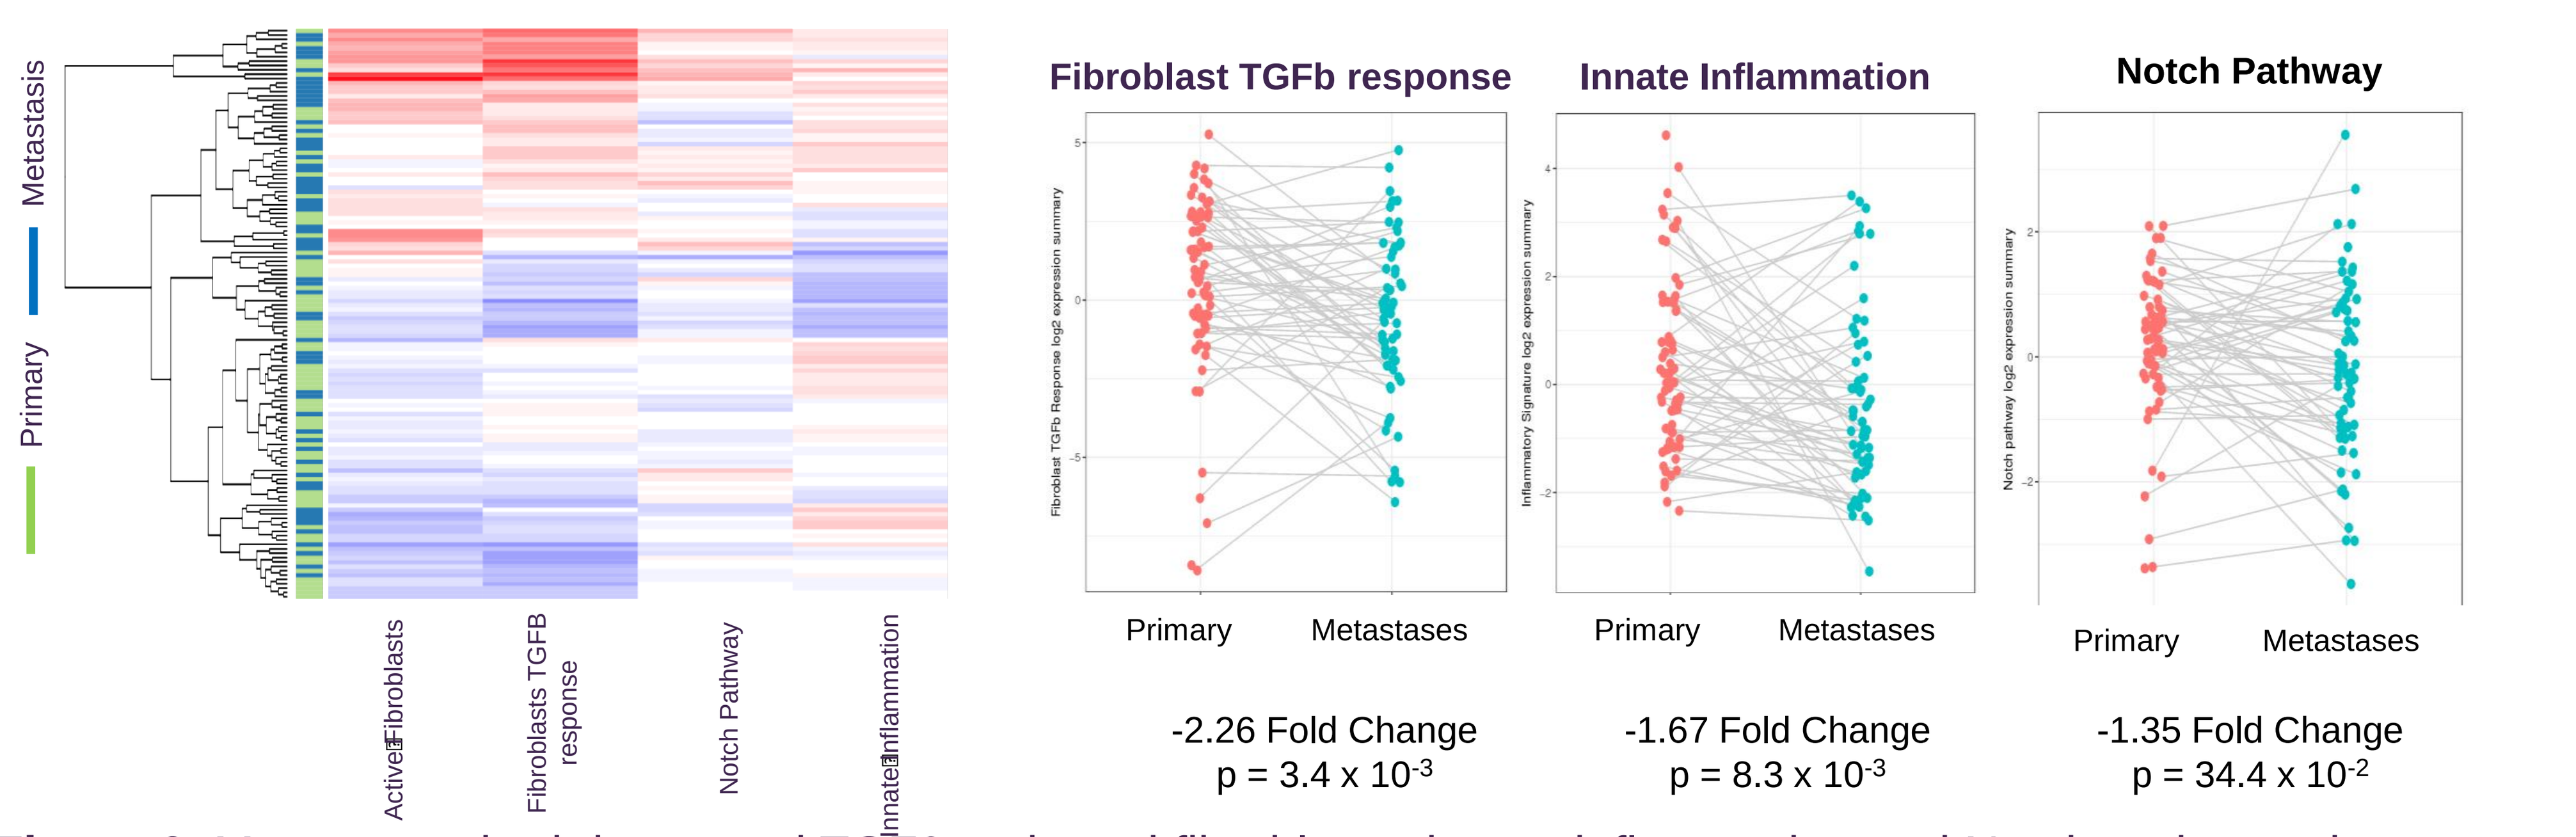
Metastasis
Primary
Active Fibroblasts
Fibroblasts TGFB response
Notch Pathway
Innate Inflammation
Fibroblast TGFb response
Innate Inflammation
Notch Pathway
Primary
Metastases
Primary
Metastases
Primary
Metastases
-2.26 Fold Change
p = 3.4 x 10-3
-1.67 Fold Change
p = 8.3 x 10-3
-1.35 Fold Change
p = 34.4 x 10-2

Figure 6. Metastases had decreased TGFβ-activated fibroblasts, innate inflammation and Notch pathway signatures

CONCLUSIONS

- ✓ PD-L1 expression and immune gene signatures did not significantly change between primary and metastatic BC
- ✓ However, metastasis had decreased TGFb-activated fibroblasts, innate inflammation and Notch pathway signatures
- ✓ The results in the current study need to be further corroborated in an independent dataset

REFERENCES

1. Fridman et. al. *Nat Rev Clin Oncol*. (2017) Jul 25
2. Herbst et. al. *Nature* (2014). 515(7528): 563–567

ACKNOWLEDGMENTS

We thank the investigators and patients of the ConvertHER study (GEICAM/2009-03), as well as the staff of GEICAM.

This presentation is the intellectual property of the authors.
Contact at:
secretaria-cientifica@geicam.org
for questions and permission to reprint and/or distribute